Quantitative Application of Up-Front Collision-Induced Dissociation for Direct Measurement of PEGylated Compounds in Dog Plasma

Patrick Lin[1], Dale Schoener[1], Melanie Gee-Shihabi[1], Richard Goodin[2], Yuan Song[2], Lida Antonian[2]

[1]Alta Analytical Laboratory, El Dorado Hills, CA
[2]Nektar Therapeutics, Huntsville, AL and San Carlos, CA

INTRODUCTION

"Up-front" collision-induced dissociation (CID) occurs in the orifice to Q0 region of a mass spectrometer (Figure 1). In this study, up-front CID coupled with tandem mass spectrometry was used to develop an MS/MS/MS application for the quantitation of PEGylated compound x. The traditional approach involved extensive sample preparation including solid phase extraction for removal of unPEGylated compounds, hydrolysis to cleave off the PEG moiety, liquid-liquid extraction and LC/MS/MS to obtain indirect measurement of PEGylated compounds. The current up-front CID method is a novel approach providing direct measurement of PEGylated compounds with superior quantitation results.

METHODS

The pH of plasma samples was adjusted to 4.4. An automatic solid phase extraction method was developed on Tomtec Quadra 96™ work station. A Waters Oasis 96-well extraction plate was used for extraction and removing of the matrix and unPEGylated compounds. The PEGylated compounds were eluted with 1 mL of ACN solution and 5 µL of the solution was injected into a Sciex API 3000 tandem mass spectrometer. The up-front CID occurred in the high pressure orifice-Q0 region by applying a relatively high orifice voltage or declustering potential (DP) to fragment all sized PEG-X mixtures into unPEGylated fragments (see Figure 1-3). The selected unPEGylated fragment (PEG_X – PEG) was further fragmented in the Q2 region of tandem mass spectrometer to obtain MS/MS/MS spectra. The ions were monitored in the MRM mode.

RESULTS

The results from this methodology are compared to the original method for overall performance, robustness, complexity and speed. The original assay included extensive sample preparation and an internal standard could only be introduced after the hydrolysis of the PEGylated compounds (Figure 5). Therefore, the reliability of quantitation data from original method is questionable due to lack of IS tracking for SPE and hydrolysis. The current assay is simple, direct and reliable whereby sample preparation is minimal and is automated (Figure 4). A PEGylated internal standard was used for the SPE and MS/MS/MS methods. This assay provided at least a six-fold improvement in sample throughput.

The data presented highlight the advantages of up-front CID approach (Figure 6,7), and the precision and accuracy for the quantitation (Table 1). The calibration range for this discovery study was 2 – 400 µg/mL in dog plasma with 98 – 104 % of accuracy and less than 16 %CV. The extraction recovery was above 60%. The established stability data in Table 2 included refrigerator, reinjection, matrix stability at room temperature, and 3-cycle freeze / thaw stability.

Figure 1: Tandem Mass Spectrometer Ion Optics Path

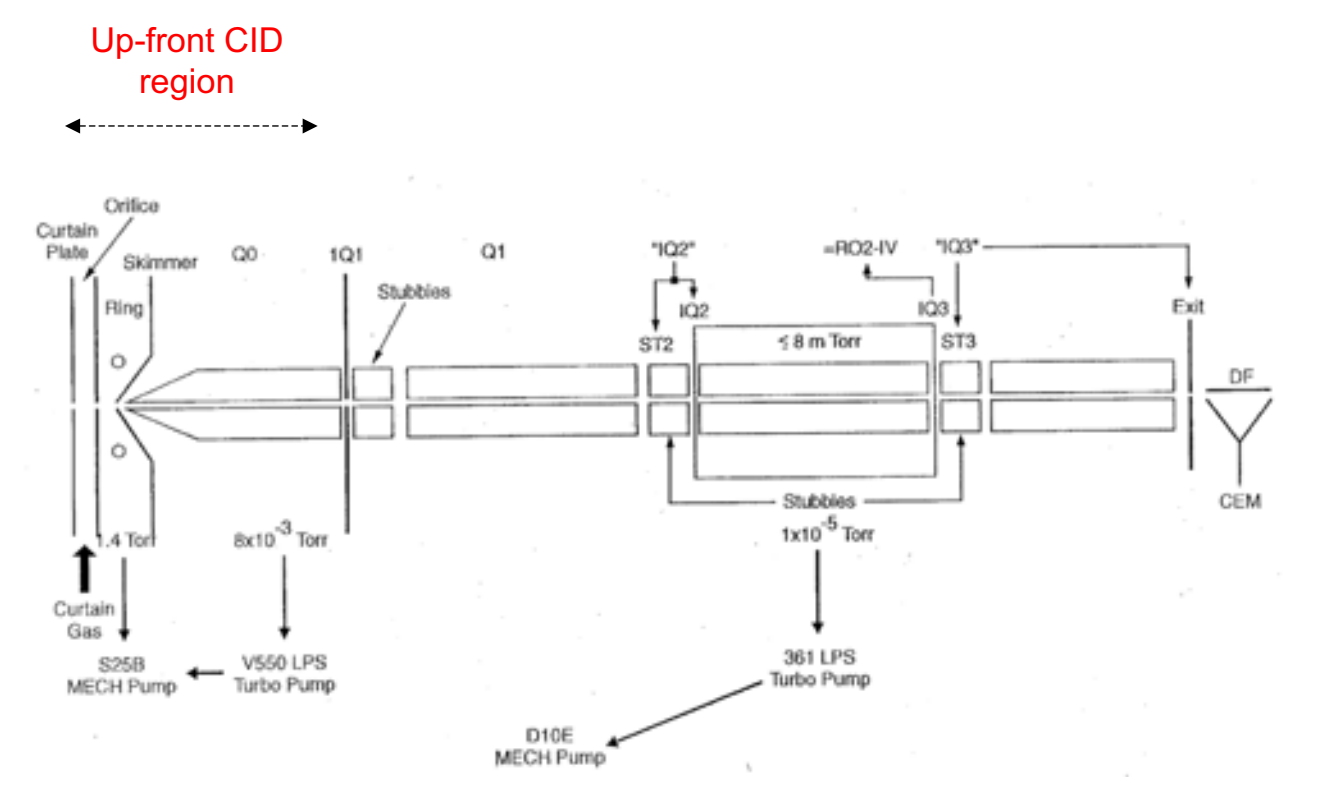
Up-front CID region
Orifice
Curtain Plate
Skimmer
Ring
Q0
IQ1
Q1
Stubbies
"IQ2"
IQ2
ST2
=RO2-IV
"IQ3"
IQ3
ST3
Exit
DF
CEM
Curtain Gas
S25B MECH Pump
V550 LPS Turbo Pump
361 LPS Turbo Pump
D10E MECH Pump

Figure 2: Q1 scan spectrum of PEG-X with normal DP value

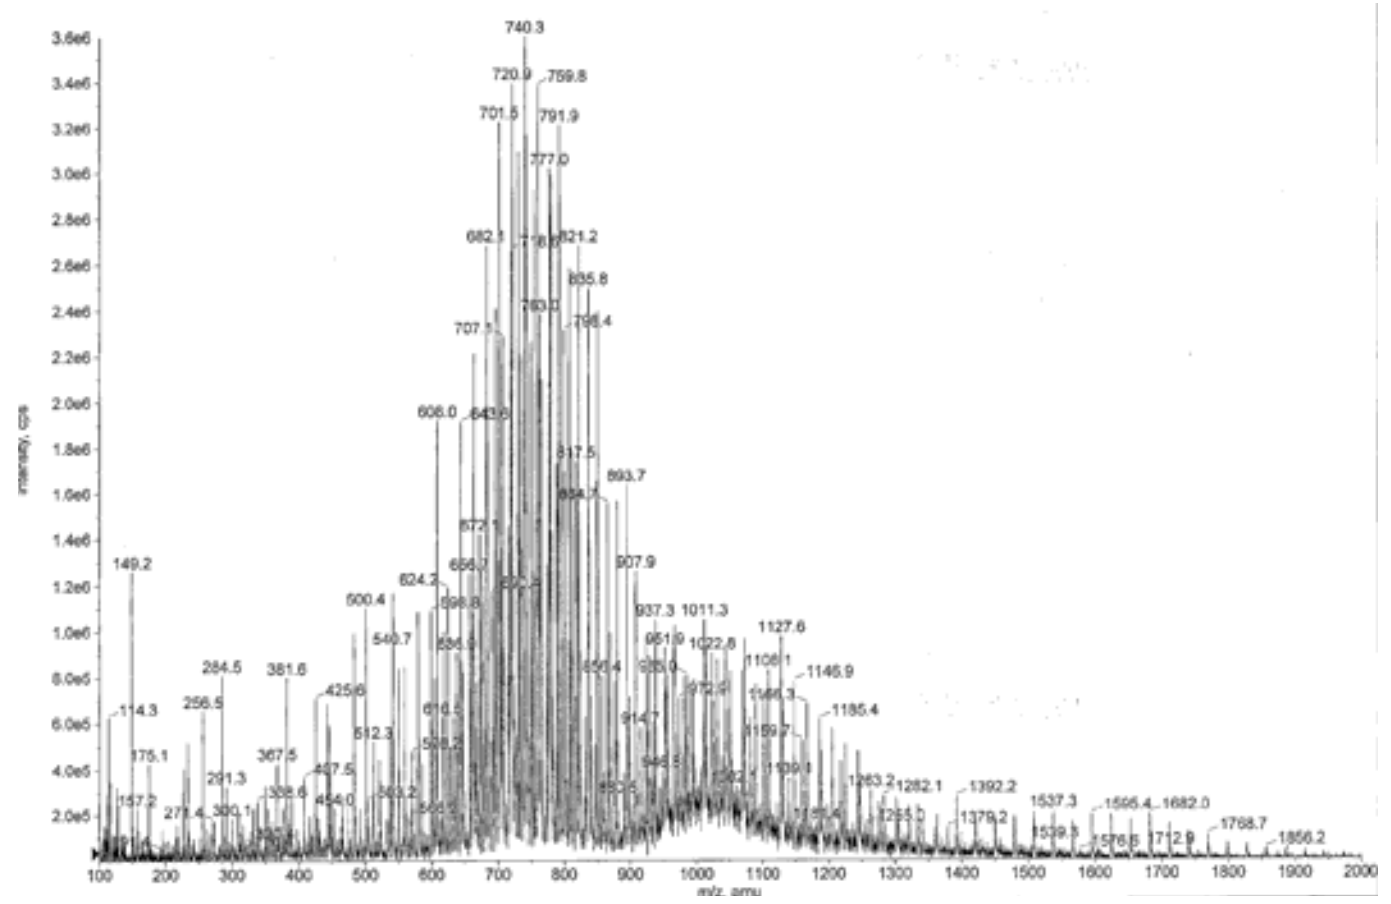
740.3
m/z, amu

Figure 3: Q1 scan spectrum of PEG-X with high DP value to produce up-front CID

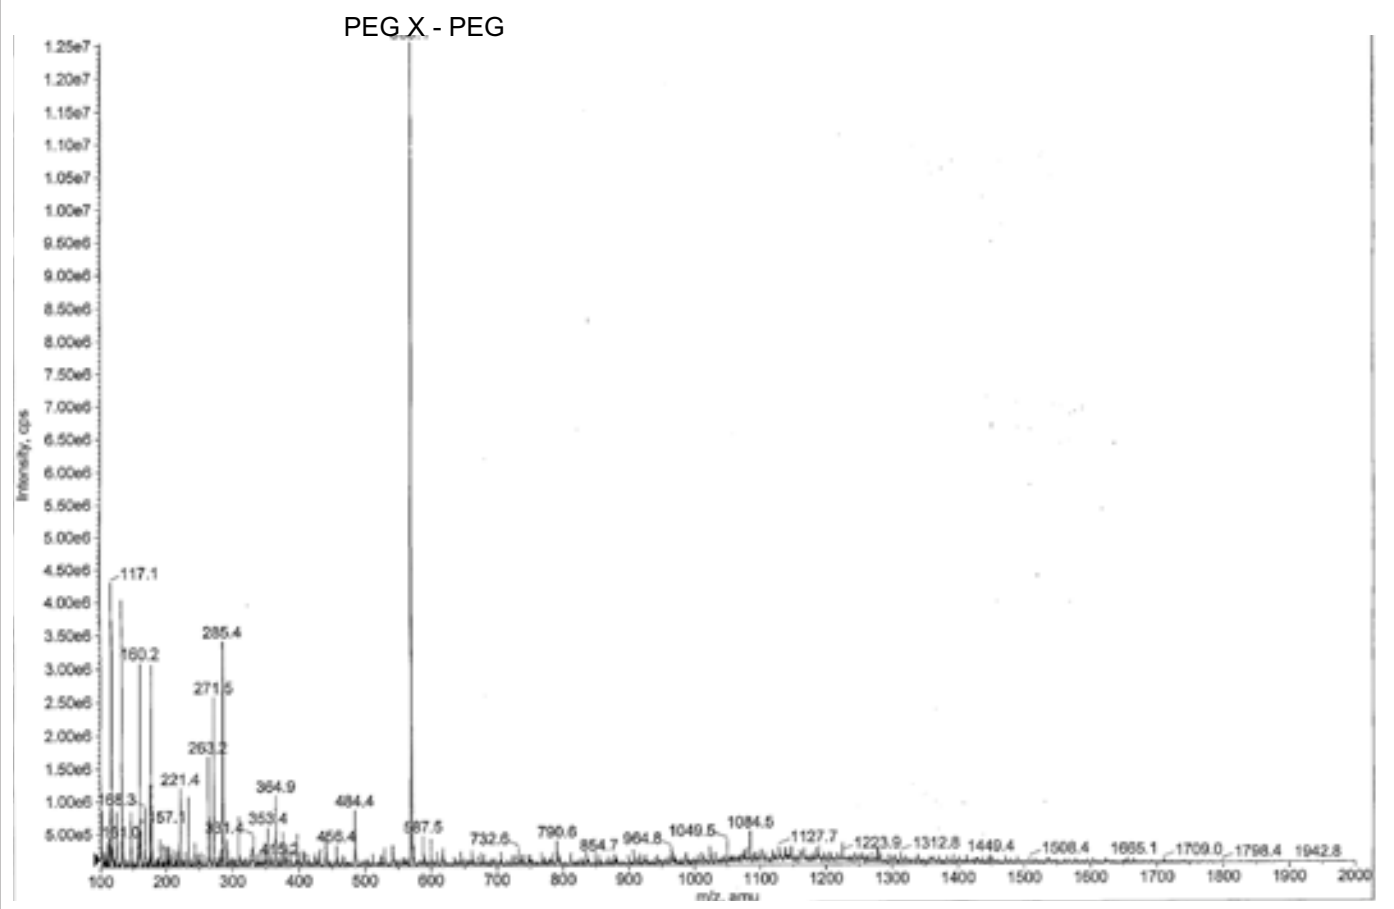
PEG X - PEG
m/z, amu

Figure 4: Sample Preparation Procedures for Direct Measurement of PEG-X

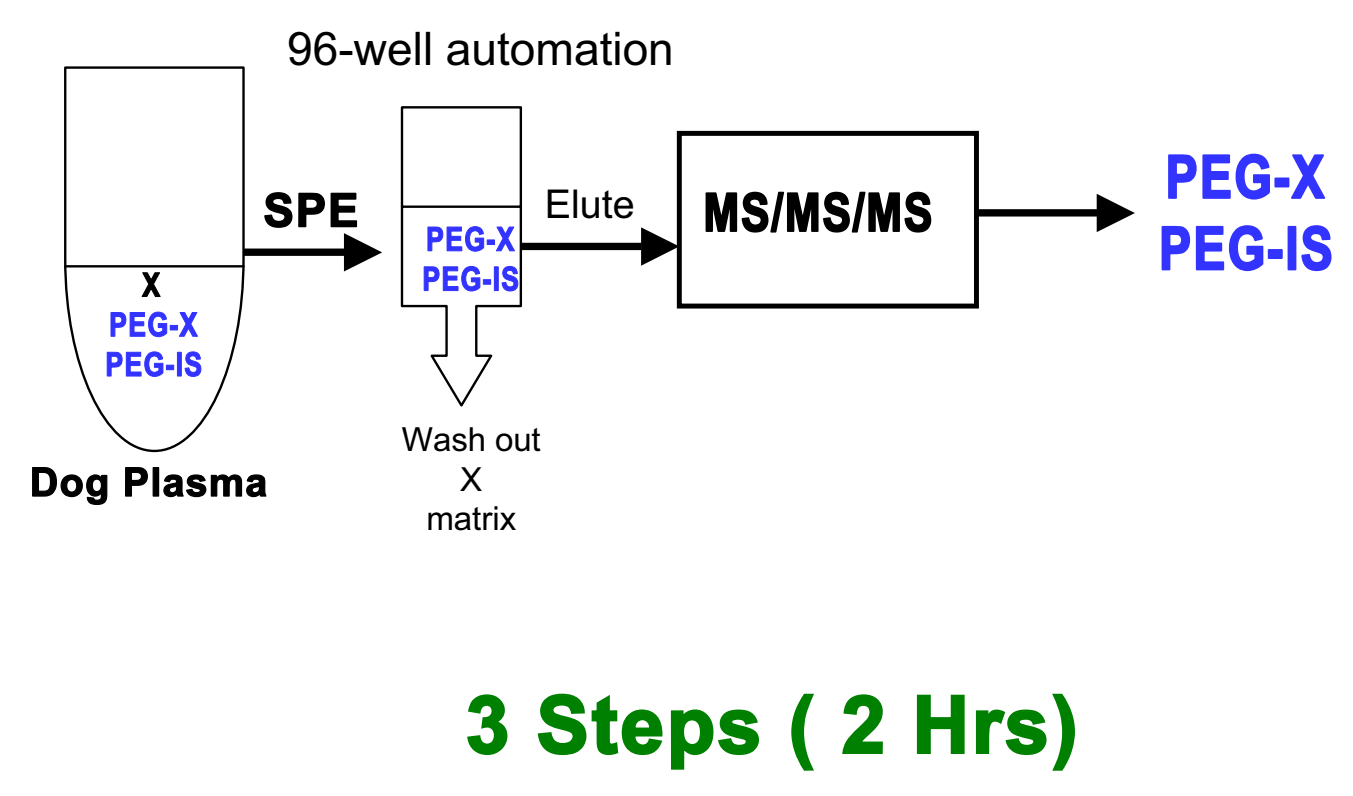
96-well automation
X
PEG-X
PEG-IS
Dog Plasma
SPE
PEG-X
PEG-IS
Wash out
X
matrix
Elute
MS/MS/MS
PEG-X
PEG-IS
3 Steps (2 Hrs)

Figure 5: Sample Preparations for Traditional Indirect LC-MS/MS Analysis of PEG-X

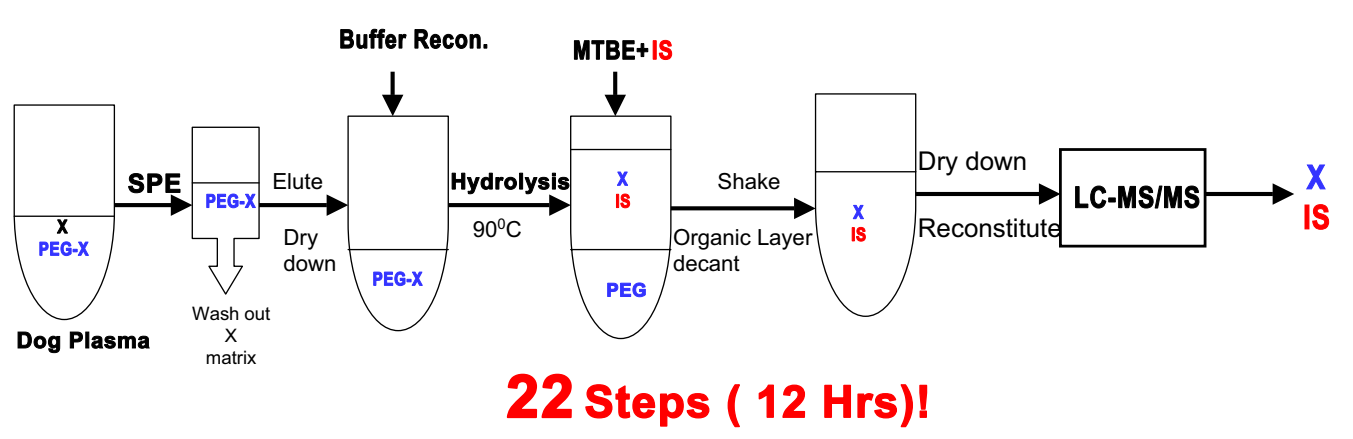
X
PEG-X
Dog Plasma
SPE
PEG-X
Wash out
X
matrix
Elute
Dry down
Buffer Recon.
PEG-X
Hydrolysis
90°C
MTBE+IS
X
IS
PEG
Shake
Organic Layer decant
X
IS
Dry down
Reconstitute
LC-MS/MS
X
IS
22 Steps (12 Hrs)!

Figure 6: MRM of Extract with Internal Standard only: PEG-X (left), PEG-IS (right)

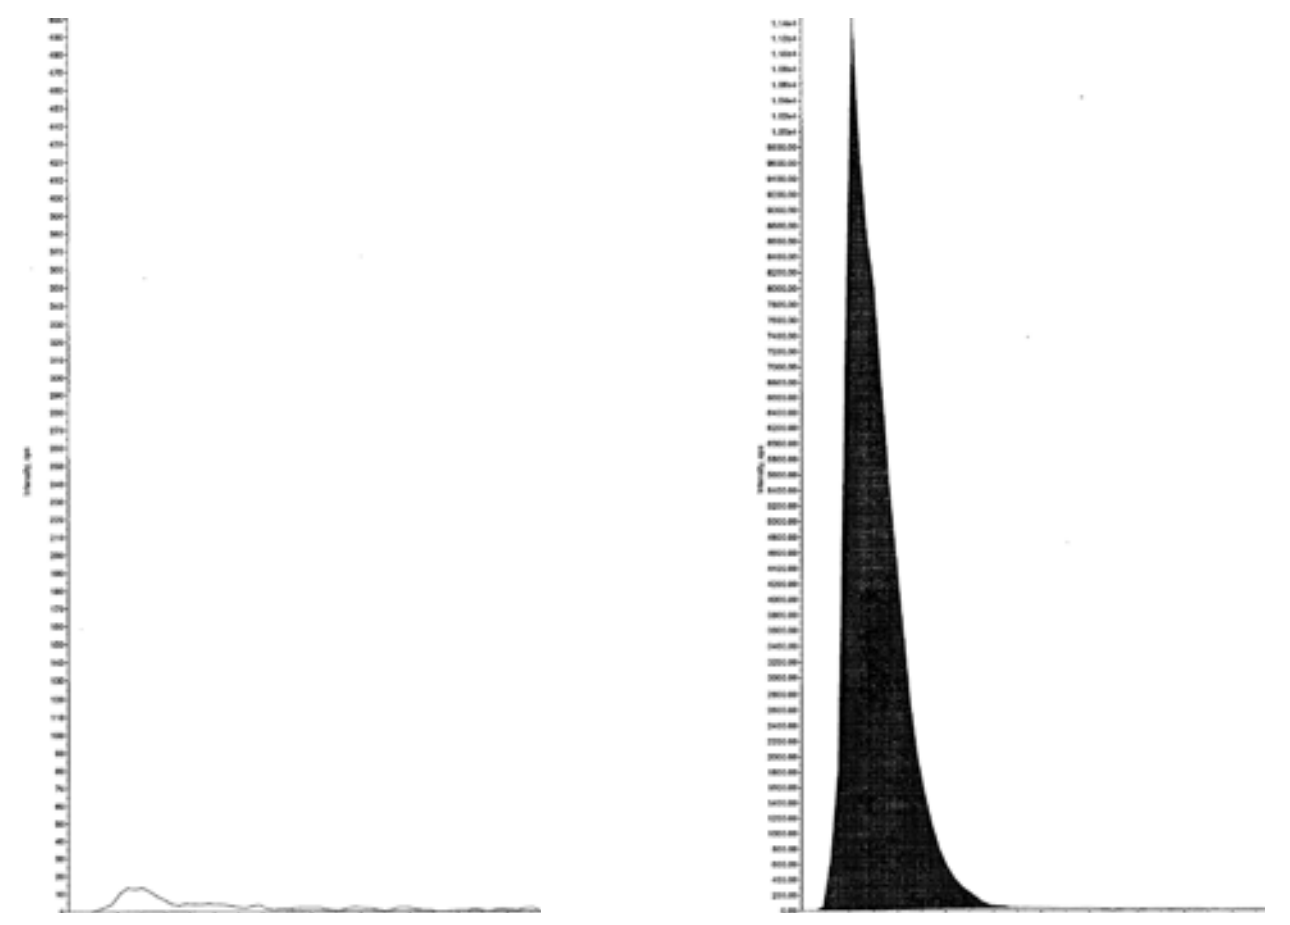

Figure 7: MRM of LLOQ Extract: PEG-X (left), PEG-IS (right)

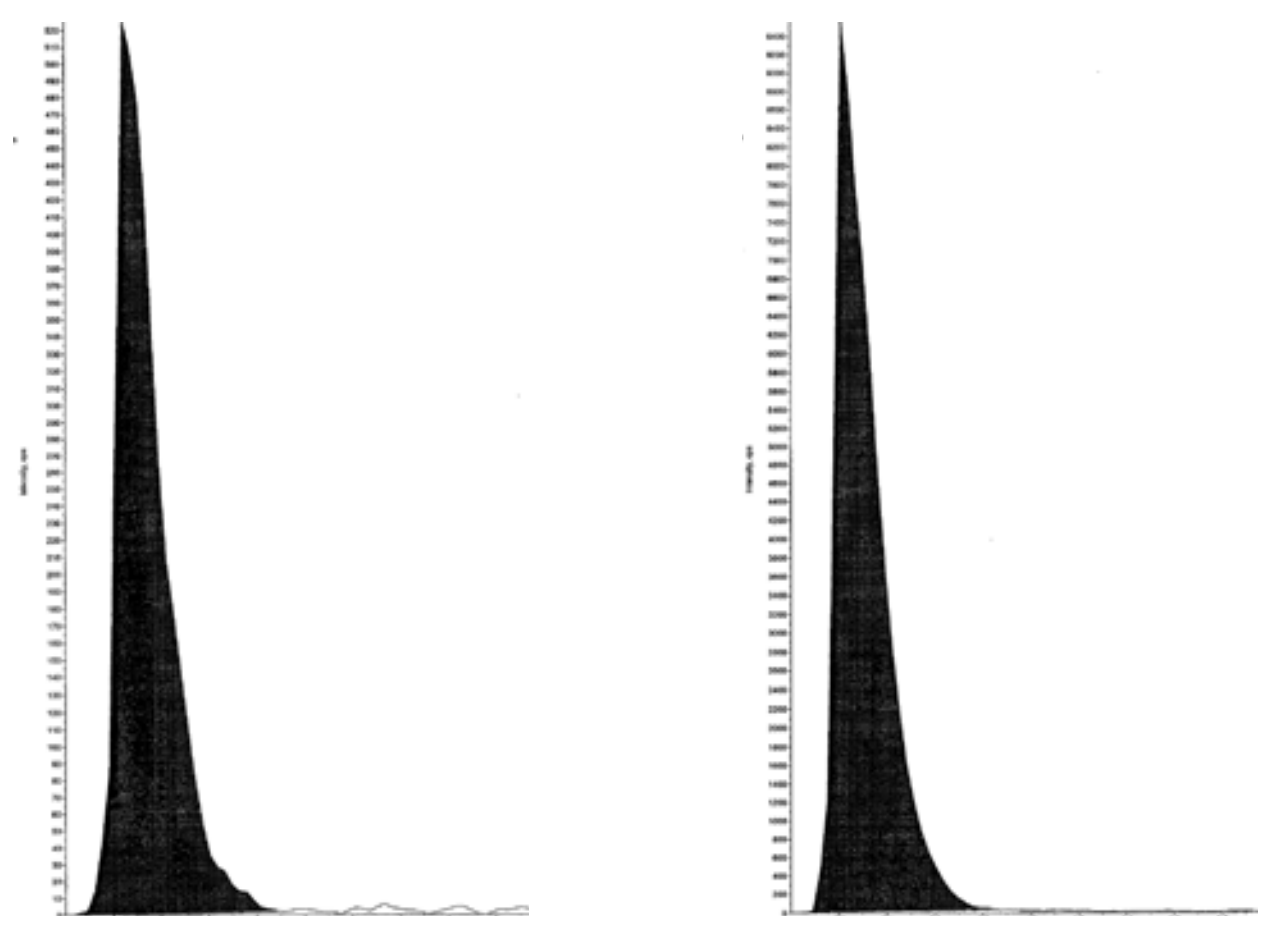

Table 1: Quality Control Statistics for PEG-X in Dog Plasma

Run #		Quality Control Concentrations (µg/mL)		
		6.00	**100**	**280**
1	Mean	5.40	98.3	277
	sd	0.732	3.15	13.5
	%CV	13.6	3.21	4.86
	%Acc.	90.0	98.3	98.9
2	Mean	5.41	95.0	272
	sd	0.507	2.94	15.7
	%CV	9.37	3.09	5.79
	%Acc.	90.2	95.0	97.1
3	Mean	6.89	111	298
	sd	0.828	8.93	21.1
	%CV	12.0	8.06	7.08
	%Acc.	115	111	106
Between - Batch	Mean	5.87	101	282
	sd	0.969	8.58	19.3
	%CV	16.5	8.48	6.83
	%Acc.	97.8	101	101

Table 2: Summary of Validation Results for PEG-X in Dog Plasma

Assay Range	2.00-400 µg/mL		
Extraction Efficiency of Analyte	74.0-78.4%		
Extraction Efficiency of Internal Standard	69.4%		
		%CV	%Accuracy
LLOQ Determination	2.00 ug/mL	8.12	114
Dilution integrity	1:10, 1:20 and 1:50	2.59-6.60	94.1-97.4
Short-term stability in plasma (RT)	24 hours	3.03-5.62	91.1-98.8
Freeze/thaw stability in plasma	3 cycles	6.57-10.3	111-121
Extract stability (4°C)	26 hours	2.99-12.9	89.5-93.2
Re-injection stability (RT) re-injected curve	34 hours	4.45-6.58	94.9-110